Advair® Prescribing Patterns in a State Medicaid and Child Health Insurance Program

Mark E. Helm, MD, MBA[1]; Perla A. Vargas PhD[2]; Stacie M. Jones, MD[2]
[1]Department of Pharmacy Practice, College of Pharmacy and Evidence-based Prescription Drug Program (EBRx)
[2]Atkansas Children's Hospital Research Institute and Department of Pediatrics, College of Medicine
University of Arkansas for Medical Sciences, Little Rock, Arkansas

J Allergy Clin Immunol. 2007; 119: S170

Background and Objective

Advair® is a combination inhaled steroid and long-acting beta agonist (LABA). This product is appropriately used for patients with chronic obstructive pulmonary disease (COPD), and as an asthma controller. National asthma treatment guidelines recommend Advair® as an alternative treatment for patients classified with moderate to severe persistent asthma.

The Food and Drug Administration (FDA) previously issued a black box warning for Advair® due to the observed increase in deaths of asthma patients using an LABA. The FDA currently recommends not to prescribe a LABA if symptom control can be achieved with inhaled steroids alone.

Advair® has become the most widely used inhaled asthma controller in recent years. This broad use implies that Advair® is prescribed as first line therapy despite current treatment guidelines and newer safety advice.

Our purpose was to describe treatment patterns of asthma patients beginning Advair® therapy. This analysis informed deliberations on policies to bring Advair® prescribing and use closer to treatment guidelines.

Methods

Advair® treated patients in the Arkansas Medicaid and State Child Health Insurance Programs were identified using administrative claims data. Asthma treatment and physician visit data were collected for all children and adults not covered by Medicare, not needing long-term care benefits, and continuously enrolled from 07/04 to 4/06.

Respiratory diagnoses were extracted for all recipients. Patients with COPD were identified and excluded.

Advair® is a registered trademark of GlaxoSmithKline (GSK). This research was conducted with no outside funding.

Methods (cont.)

Treatment patterns were examined for patients with new Advair® claims in 2005, who did not have a COPD diagnosis. Pharmacy claims for Advair,® inhaled asthma controllers, rescue medications, and systemic steroids were examined. Additionally, outpatient services and hospital admissions were also reviewed.

Results

Slightly more than 6.5% of the continuously eligible recipient population (n=276,521) filled at least one prescription for an inhaled asthma controller in the study period. Approximately half of patients using an asthma controller at least once, filled a prescription for Advair®. The following table categorizes recipient counts in 2005.

Recipient Category in Year 2005	Count	%
Total population using Advair	8162	
Advair users with COPD diagnosis	1429	18%
Advair Users with Asthma (79% with asthma code, 21% with no respiratory diagnosis)	6733	82%
Prior Advair users with asthma (filling at least one Advair claim from 7/04 to 12/04)	2347	29%
New Advair Users with asthma	4386	54%

Prior inhaled controller use was assessed for all asthma patients started on Advair® from January 2005 through April 2006 (n=5017). Prior use was assessed from July 1, 2004 to the date of the 2005 index Advair® claim. A small number of new Advair® patients (n=563) were treated with an inhaled steroid controller prior to starting Advair. Only 16 patients used an add-on LABA prior to their first Advair® prescription claim.

Results

Months of Inhaled Controller Use Before Advair®

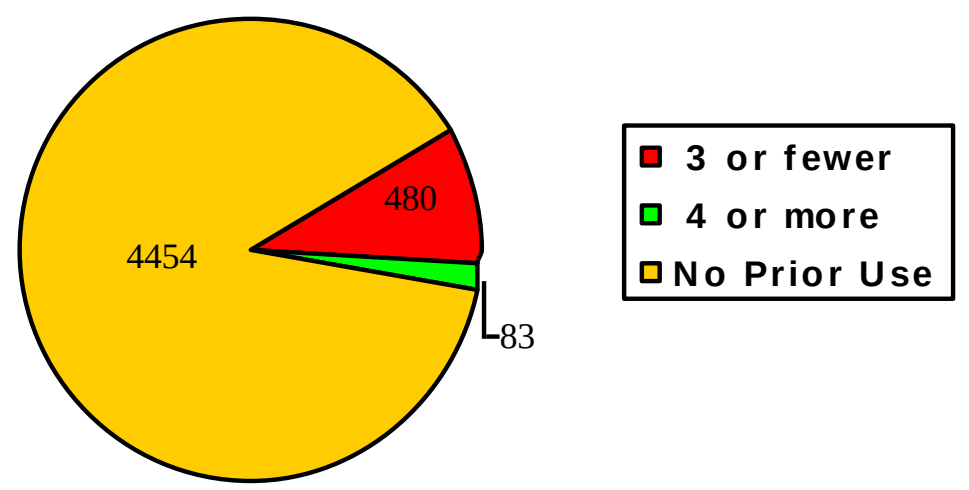
480
4454
83
3 or fewer
4 or more
No Prior Use

Short-acting Beta agonist and systemic steroid use are considered markers of asthma severity and symptom control. Prescription claims dispensed in the six months prior to and after Advair® start were examined for 2257 new patients starting Advair® in the first half of 2005.

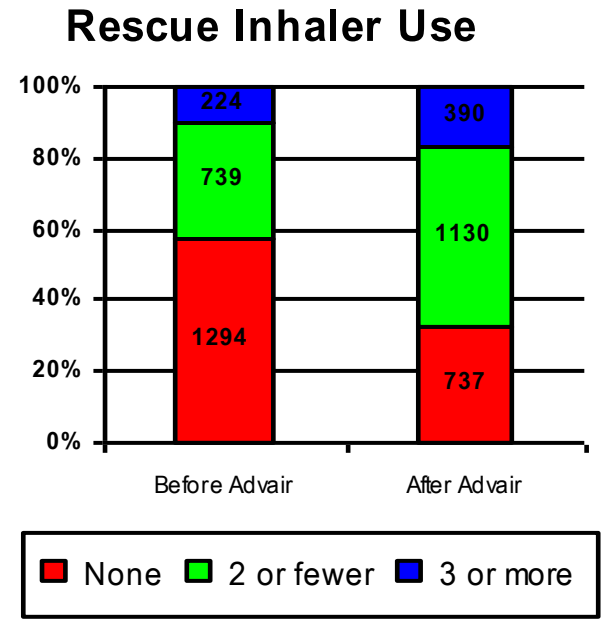
Rescue Inhaler Use
100%
80%
60%
40%
20%
0%
224
739
1294
390
1130
737
Before Advair
After Advair
None
2 or fewer
3 or more

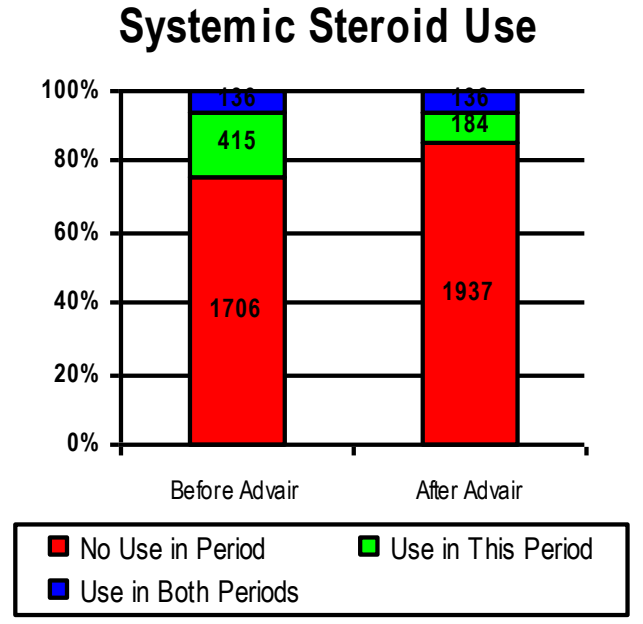
Systemic Steroid Use
100%
80%
60%
40%
20%
0%
136
415
1706
136
184
1937
Before Advair
After Advair
No Use in Period
Use in This Period
Use in Both Periods

Provider encounter patterns show that 44% of patients starting Advair® had fewer than two office visits in a year. Nearly a third of patients with ED visits were seen both before and after starting Advair.®

All Cause Encounters	Office Visits	ER Visits	Hospital
Only Before	433	333	13
Both Before and After	1267	153	0
Only After	129	261	14
No Visits	429	1510	2230

More than half of patients starting Advair® failed to obtain a refill in the subsequent six months. In the first half of 2005, only 13% of the 2257 new Advair® non-COPD patients filled three or more prescriptions within six months. Only 18 patients of the 2257 new starts filled Advair® monthly.

Advair Refills Six Months After Index Advair Claim

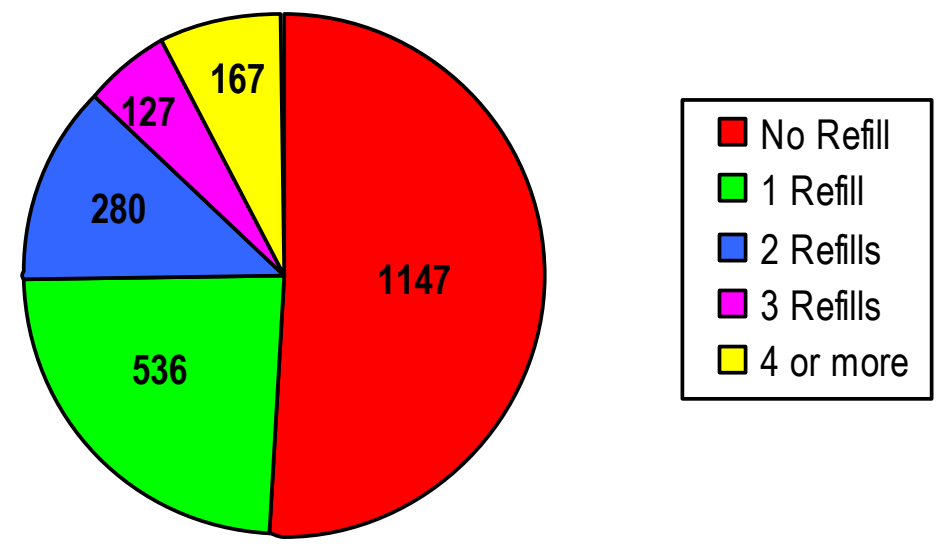
1147
536
280
127
167
No Refill
1 Refill
2 Refills
3 Refills
4 or more

Conclusions

Most patients initiating Advair® treatment in the first half of 2005 lacked markers of moderate to severe persistent asthma. The fact that 30% of patients have no claim for a rescue inhaler in combination to the sporadic refilled pattern of Advair® suggests that it may be used for treatment of exacerbations.

This pattern of use does not follow treatment guidelines or safety recommendations from the FDA and GSK. Arkansas Medicaid and SCHIP recipients may be at increased risk for life-threatening events because of Advair® use outside of accepted recommendations.

Given the apparent inappropriate use and higher risks and costs of Advair® relative to other asthma controllers, the Arkansas Medicaid program instituted Advair® prior authorization requirements for patients who lack markers of moderate or severe asthma, or who do not show a refill pattern suggestive of adherence to controller therapy.